Acrylamide Perturbs Calcium and Actin Cytoskeleton Signaling in Rat and Mouse Cancer Target Tissues (Abstract # 1830)

Chepelev NL[1], Gagné R[2], Maynor T[1], Hobbs CA[1], Kuo B[2], Yauk CL[2], Recio L[1]

[1]Integrated Laboratory Systems Inc., Research Triangle Park, NC, [2]Environmental Health Science and Research Bureau, Health Canada, Ottawa, ON, Canada

Abstract

Acrylamide (AA) exposure leads to thyroid tumors in male F344 rats, and lung and Harderian gland tumors in male B6C3F1 mice, in 2-year cancer bioassays. We used RNA-sequencing to explore the mechanisms contributing to acrylamide-induced cancers in these rodents. Male F344/DuCrl rats or male B6C3F1 mice were administered up to 24 mg acrylamide/kg bw-day AA through drinking water in five dose groups alongside controls for 5, 15, or 31 days. Thyroid glands from rats, and lungs and Harderian glands from mice were harvested. Extracted mRNA was sequenced at 10-20 million reads per sample by poly-A RNA-sequencing on an Ion Proton sequencer. Ingenuity Pathway Analysis was used to explore the molecular perturbations induced by AA and transcriptional benchmark doses (BMDs) were generated in BMDExpress.

Differentially expressed genes in both species provided marginal support for the involvement of hormonal perturbations and DNA damage in the carcinogenic mode of action (MOA) of AA. This was consistent with a lack of induction of red blood cell micronuclei, equivocal results in the Pig-a mutation assay, and marginal changes in serum thyroid hormone levels. Instead, there was a pronounced effect on calcium signaling/cytoskeletal genes (e.g., actins, myosins, troponins) in both species. Strikingly, for each tissue, calcium/actin cytoskeleton signaling pathways were the top pathways affected by AA. Calcium signaling genes affected in all rodent tissues were also altered in some human tumors (e.g., human thyroid follicular carcinoma), including Parvalbumin (PVALB), a marker of malignant thyroid tumors in humans. The lowest pathway transcriptional BMDs were within 2-fold of the BMDs derived from analysis of cancer endpoints in the rodent bioassays.

These results demonstrate that AA perturbs calcium/actin cytoskeleton signaling across tissues and species, providing greater confidence in this as a novel MOA of AA carcinogenicity in rats and mice. The proposed MOA is consistent with published effects of AA on cytoskeletal proteins, and with well-documented perturbations of calcium levels in malignant rodent and human tumors. Finally, good concordance between cancer and transcriptomic BMDs, and insight into human relevance of these findings, support the utility of toxicogenomics for human health risk assessment. This work was funded by SNF SAS and Health Canada funds.

Objectives

1) to examine the temporal-concordance and dose-response of the transcriptional data in support of genotoxicity, or alternatively, CS as a key event, in the MOA for AA-induced carcinogenicity (referred to as the genotoxicity and CS MOA analyses, respectively); **2)** to perform dose-response modeling of apical (cancer) data, as a part of the weight of evidence analysis, to compare it to transcriptional data, thus assessing the plausibility of the genotoxicity vs. CS-based MOA; and **3)** to derive a toxicogenomics-based POD for potential use in human health risk assessment of AA.

Materials & Methods

- Animals were dosed with AA in drinking water, as described in Figure below:

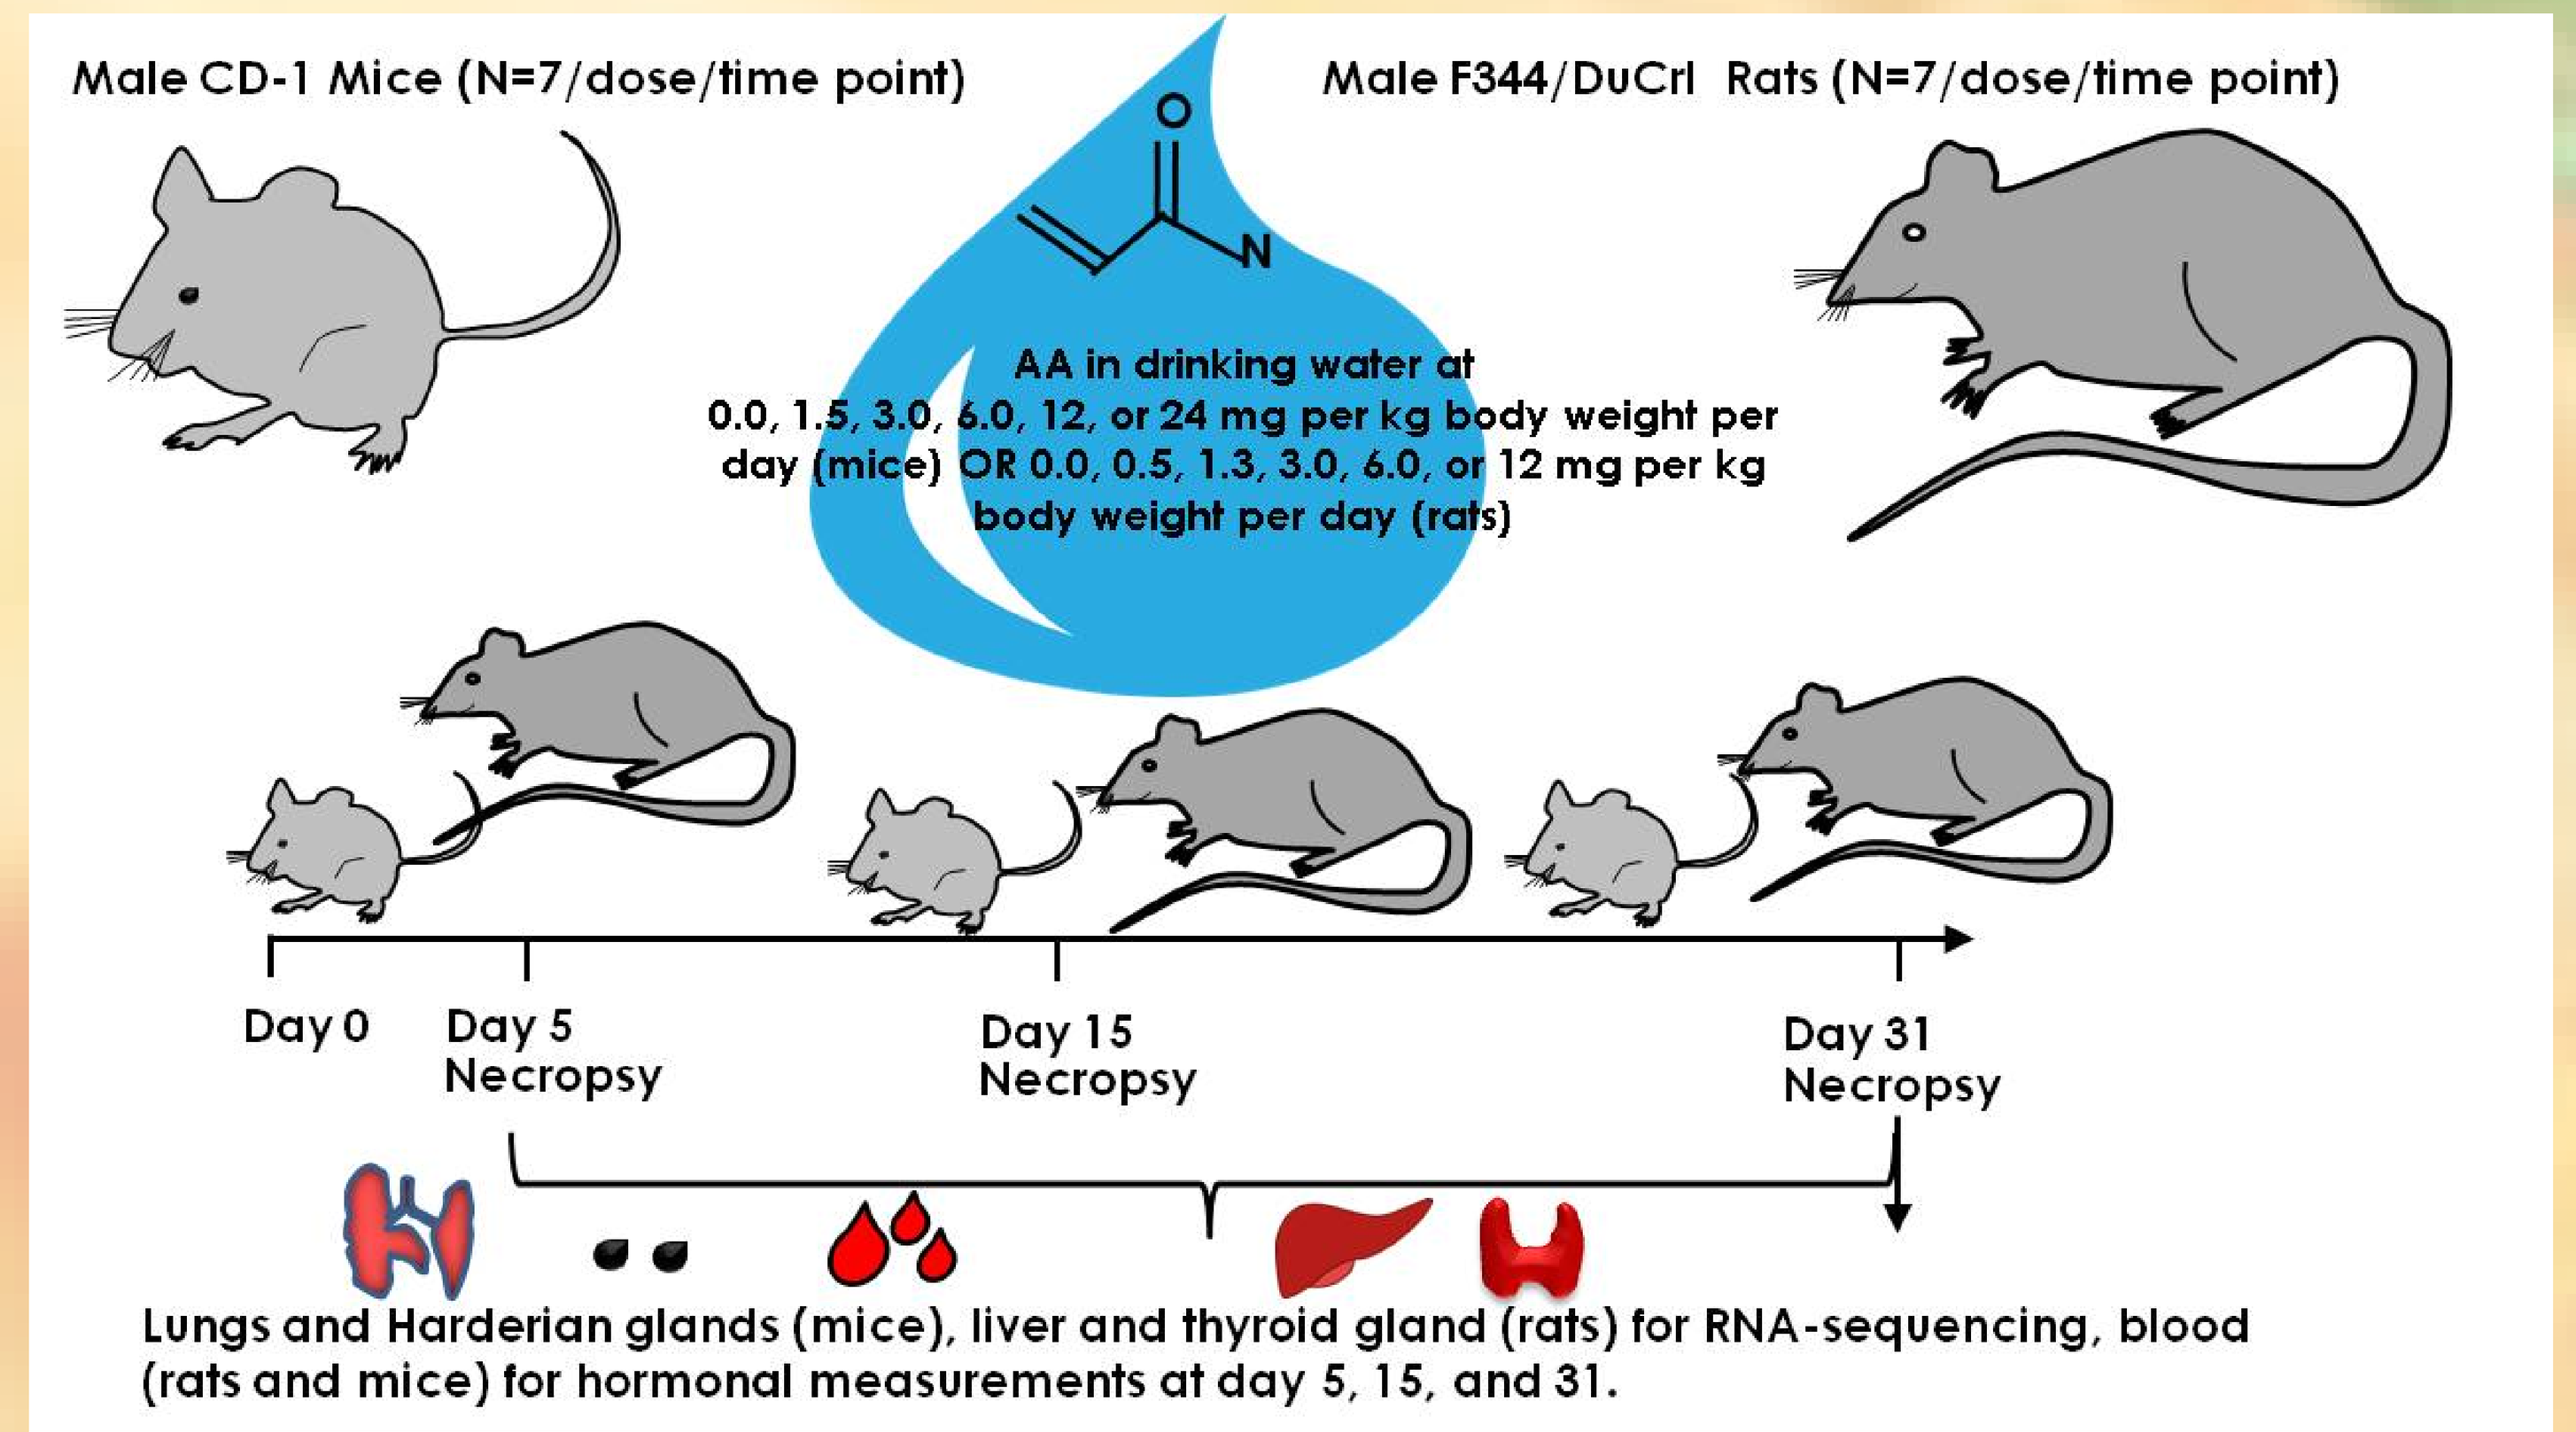
Male CD-1 Mice (N=7/dose/time point)
Male F344/DuCrl Rats (N=7/dose/time point)
O
N
AA in drinking water at 0.0, 1.5, 3.0, 6.0, 12, or 24 mg per kg body weight per day (mice) OR 0.0, 0.5, 1.3, 3.0, 6.0, or 12 mg per kg body weight per day (rats)
Day 0
Day 5 Necropsy
Day 15 Necropsy
Day 31 Necropsy
Lungs and Harderian glands (mice), liver and thyroid gland (rats) for RNA-sequencing, blood (rats and mice) for hormonal measurements at day 5, 15, and 31.

- mRNA was sequenced (20 million reads per sample) by poly-A RNA-sequencing on an Ion Proton (workflow shown below) or Illumina HiSeq sequencer;

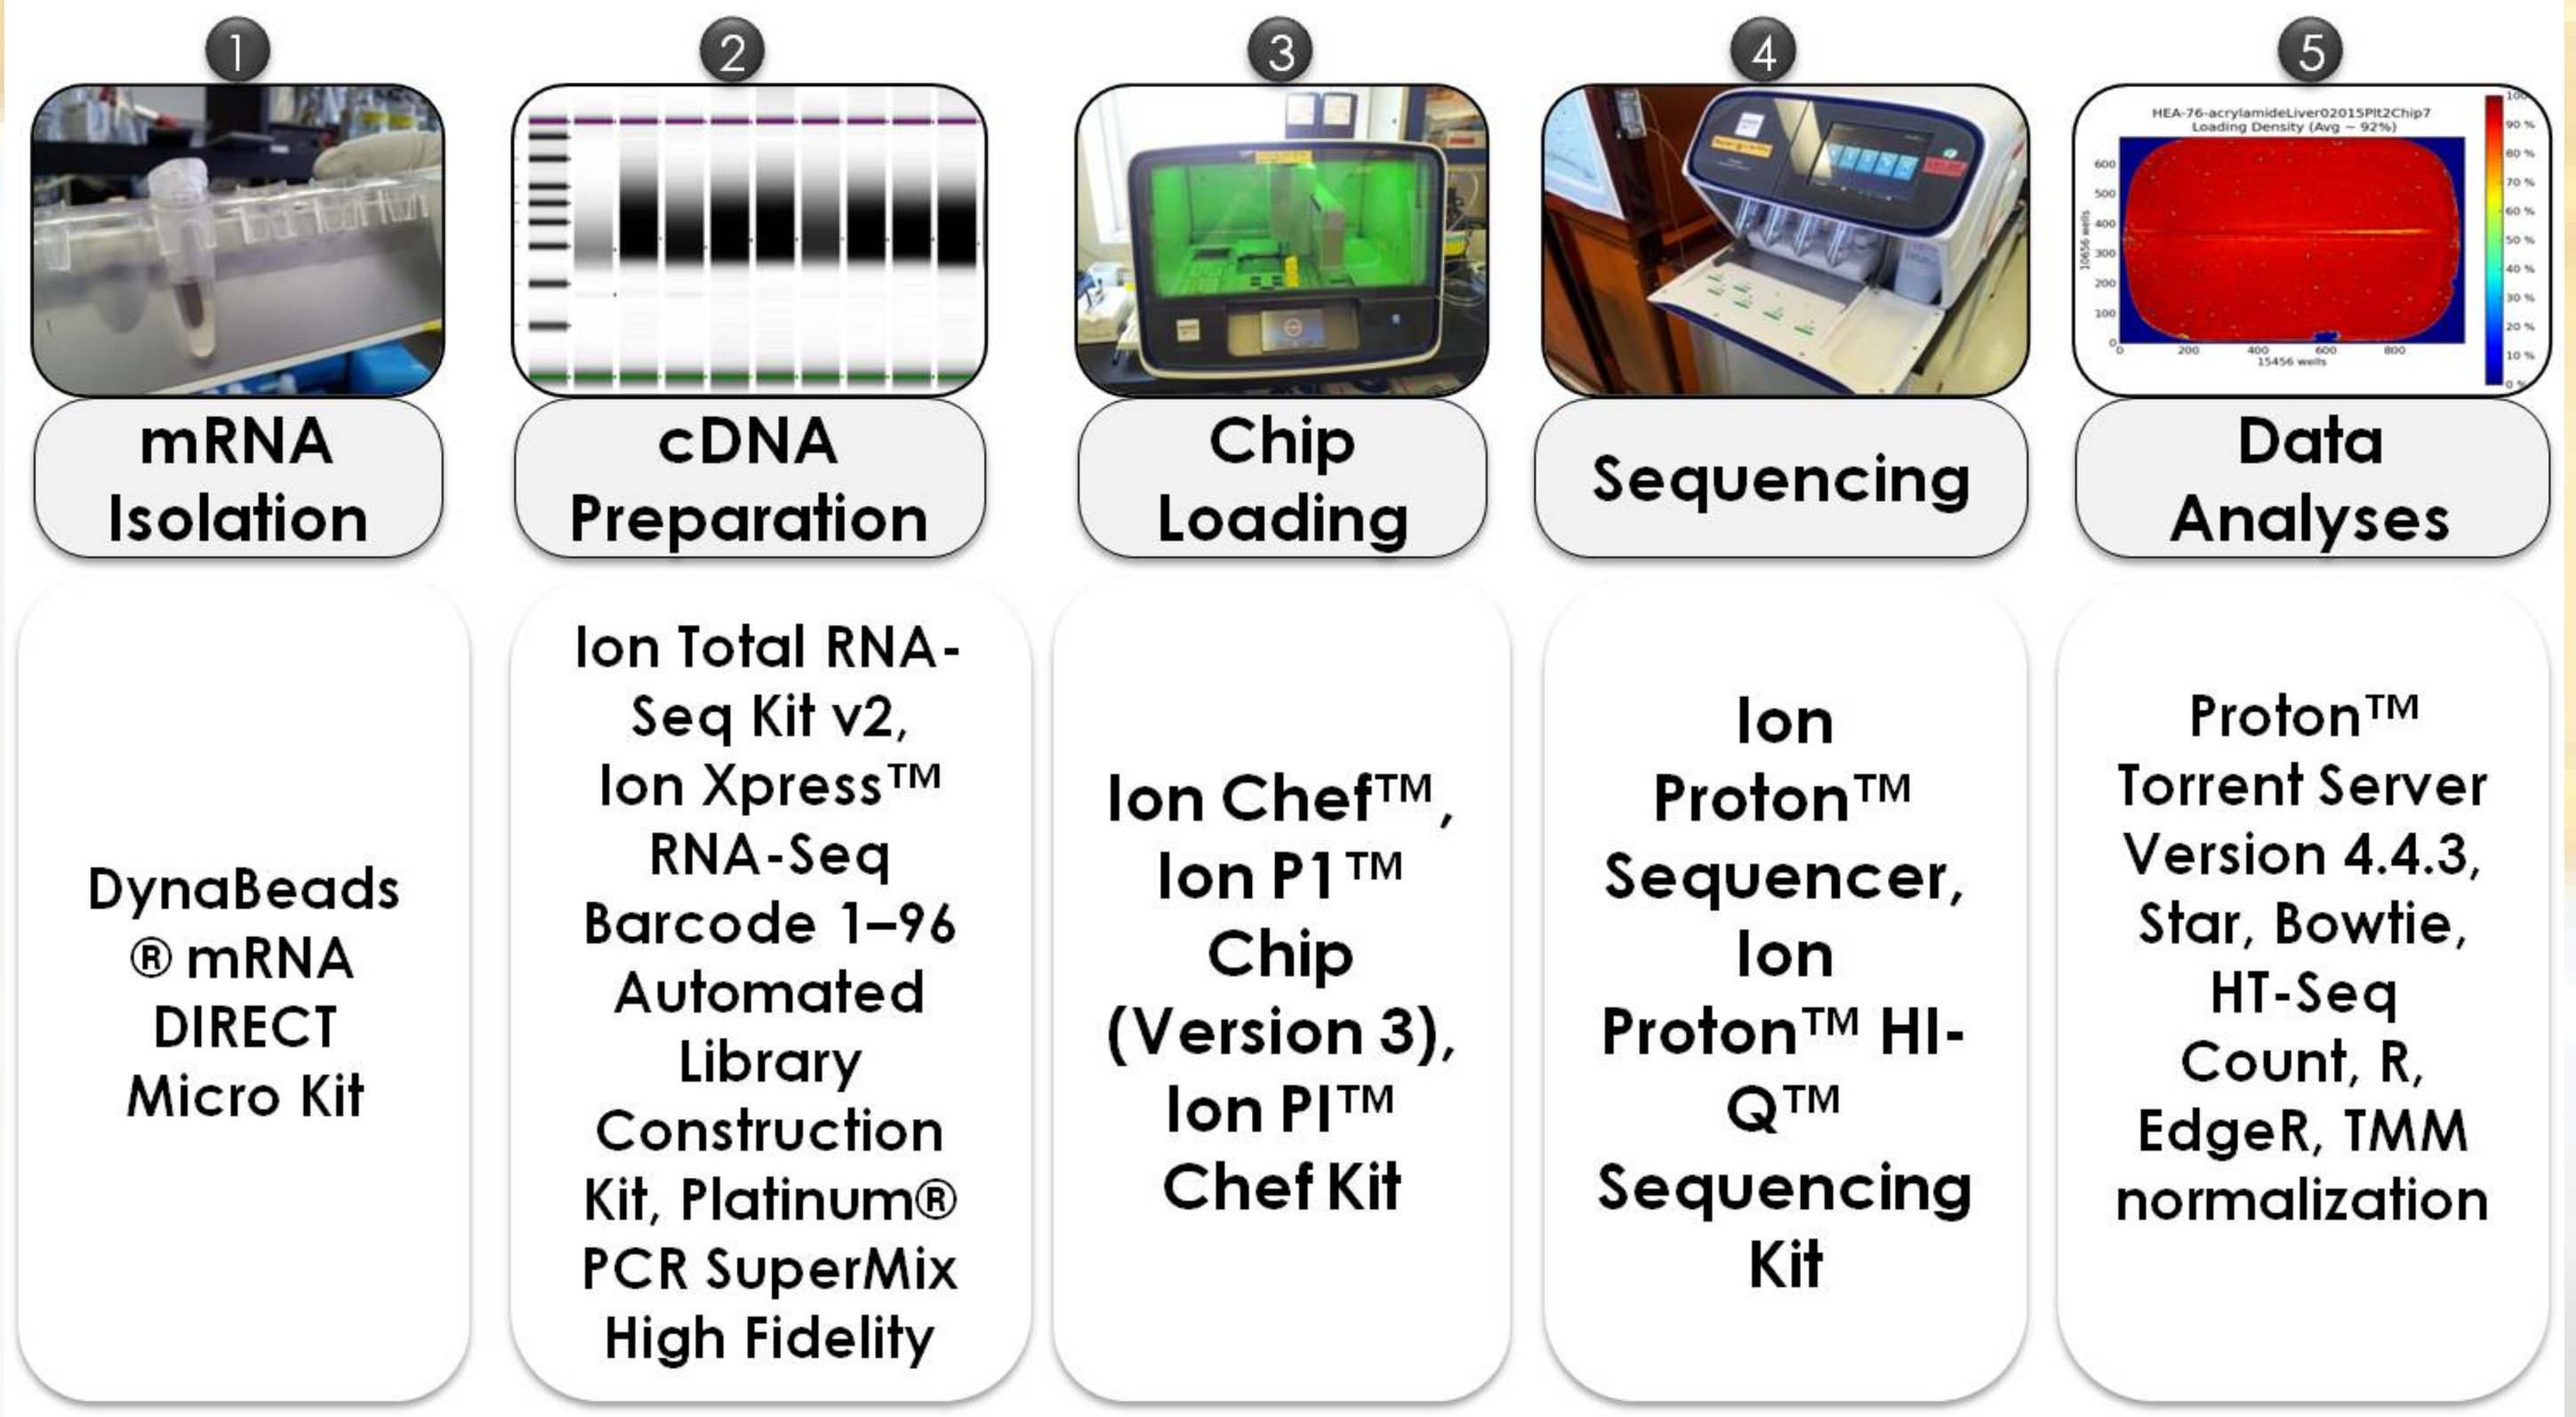
1
mRNA Isolation
DynaBeads® mRNA DIRECT Micro Kit
2
cDNA Preparation
Ion Total RNA-Seq Kit v2, Ion Xpress™ RNA-Seq Barcode 1–96 Automated Library Construction Kit, Platinum® PCR SuperMix High Fidelity
3
Chip Loading
Ion Chef™, Ion P1™ Chip (Version 3), Ion PI™ Chef Kit
4
Sequencing
Ion Proton™ Sequencer, Ion Proton™ HI-Q™ Sequencing Kit
5
Data Analyses
Proton™ Torrent Server Version 4.4.3, Star, Bowtie, HT-Seq Count, R, EdgeR, TMM normalization

- EdgeR was used for RNA-seq data analysis, with TMM normalization;
- Serum thyroid hormone levels were measured at AniLytics, Inc.;
- Blood micronucleus (MN) and *Pig-a* assays were conducted using MicroFlow® kit (Litron Laboratories).

Results

The most striking feauture was that CS or related pathways were affected by AA in the three target cancer, but not non-target tissues (Figure below):

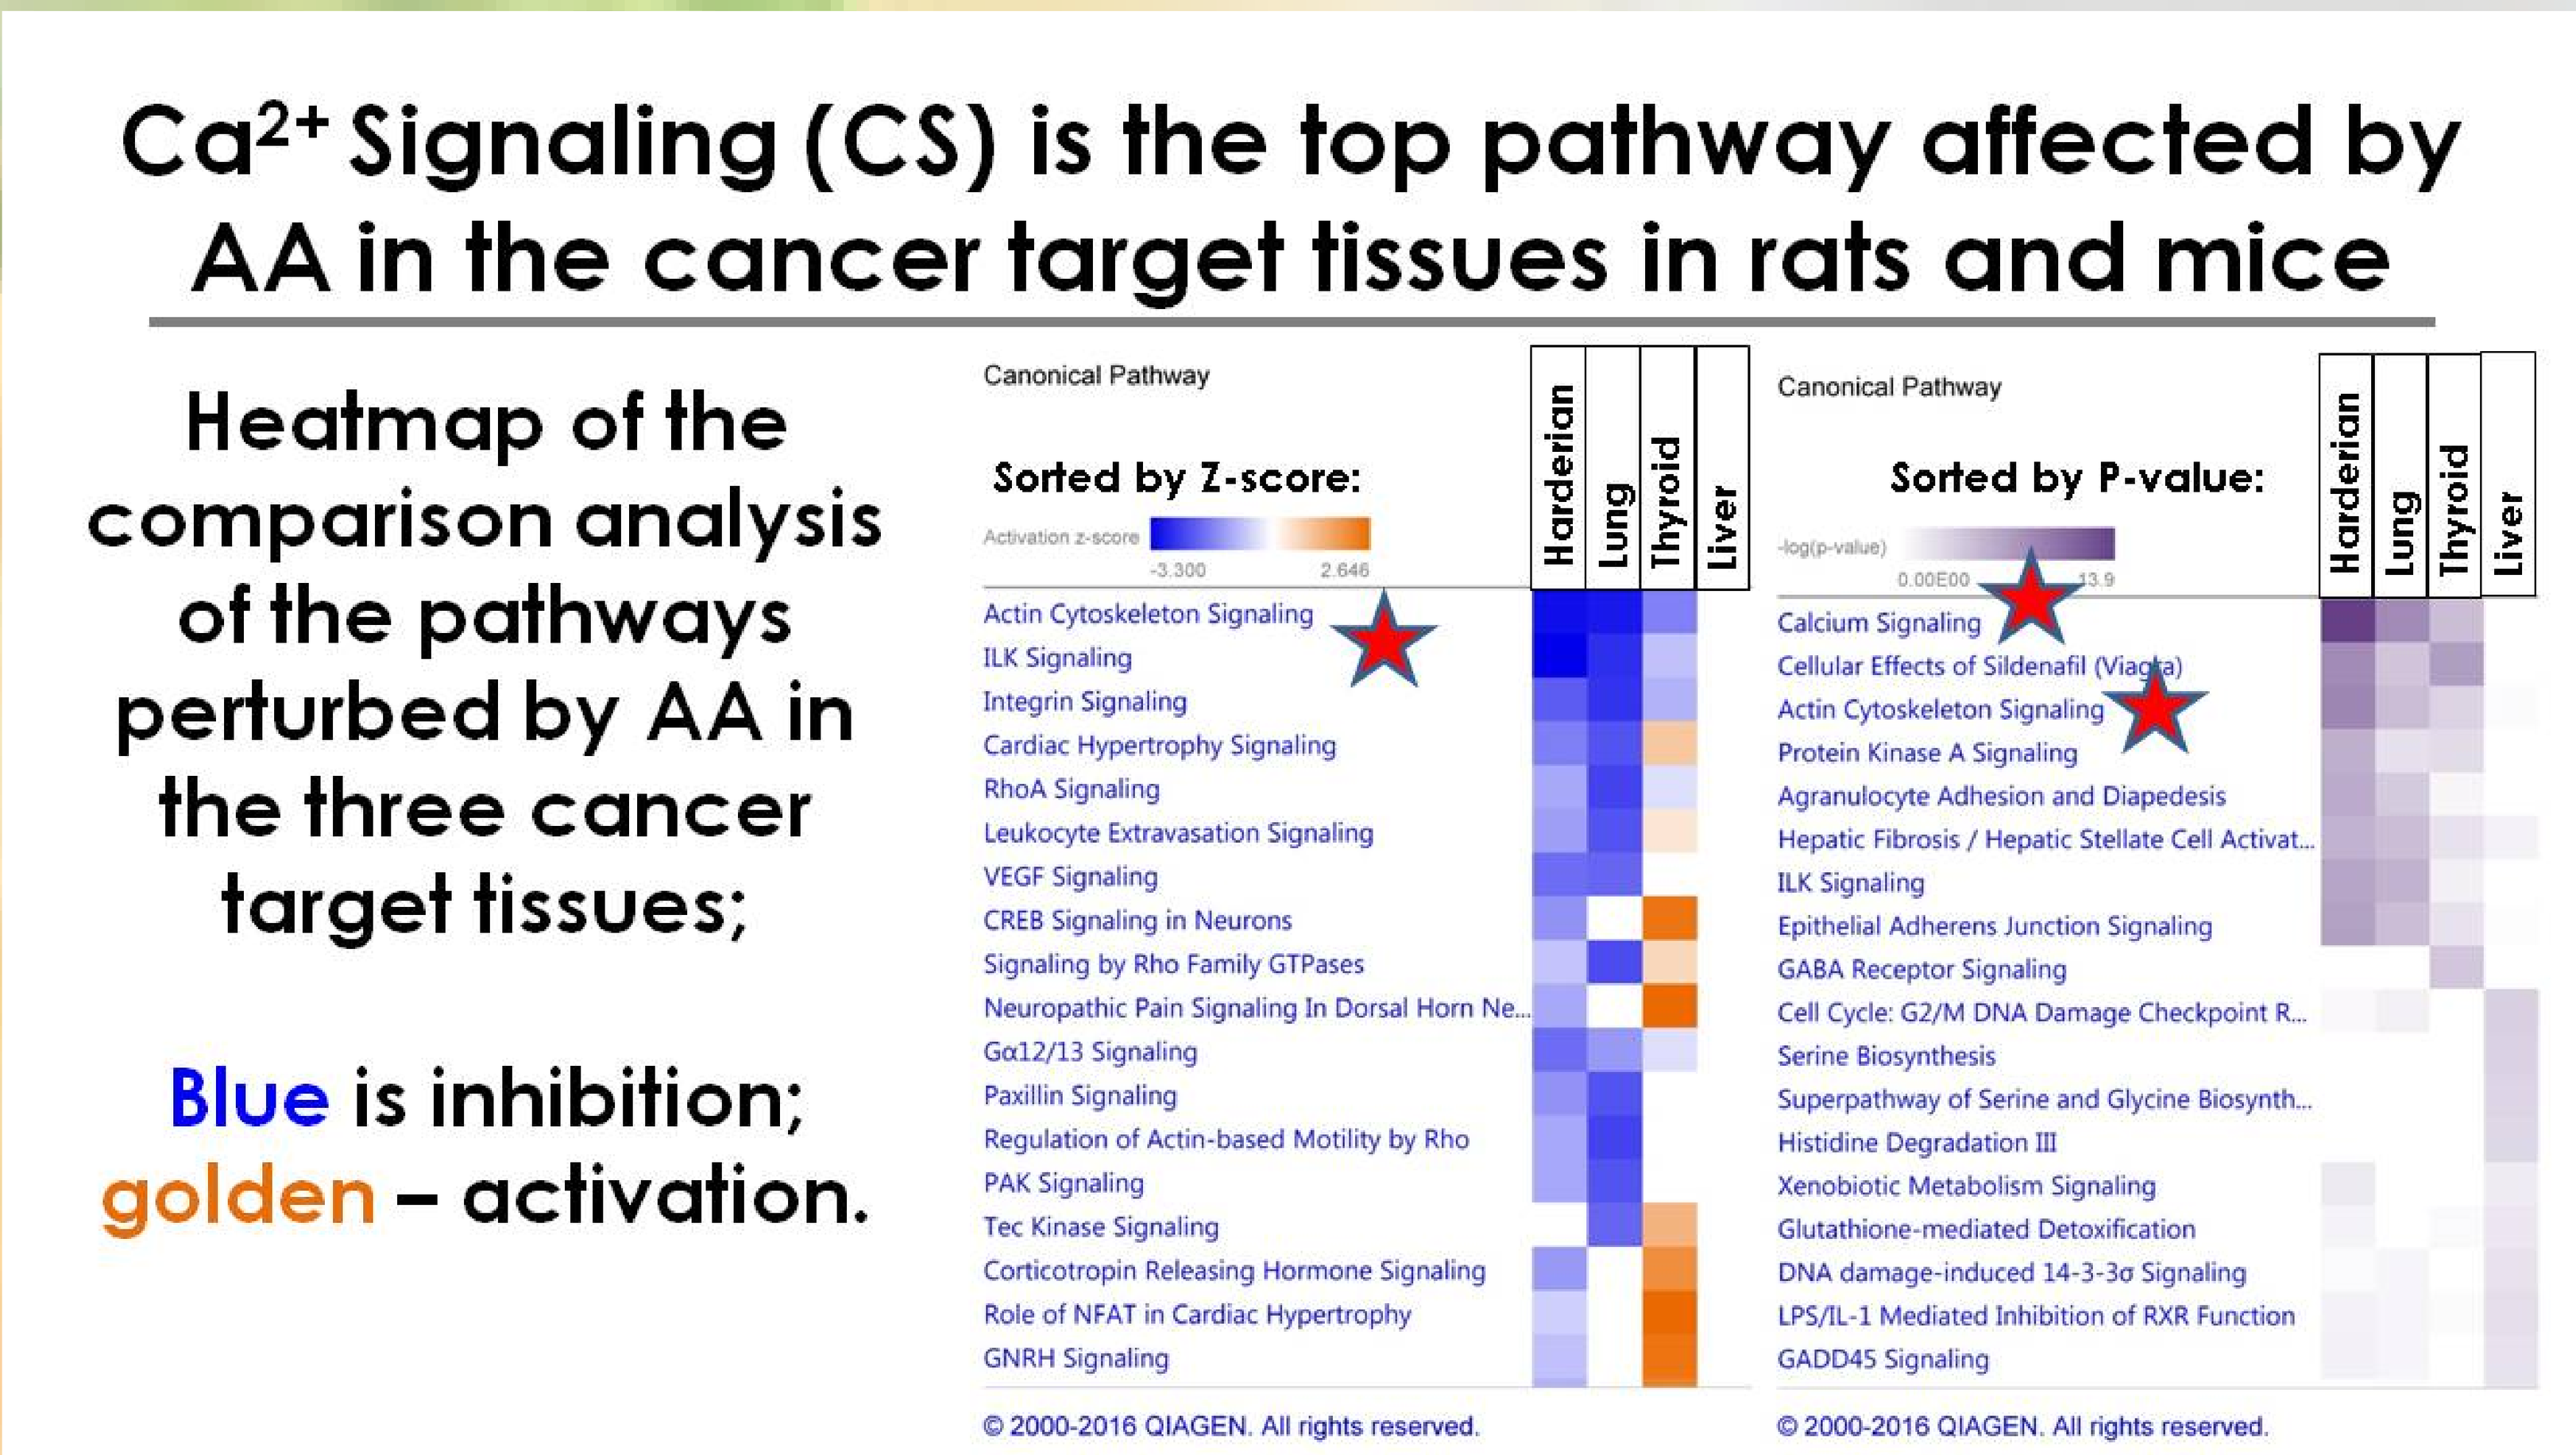
Ca^{2+} Signaling (CS) is the top pathway affected by AA in the cancer target tissues in rats and mice
Heatmap of the comparison analysis of the pathways perturbed by AA in the three cancer target tissues;
Blue is inhibition; golden – activation.
Canonical Pathway
Sorted by Z-score:
Activation z-score
-3.300
2.646
Harderian
Lung
Thyroid
Liver
Actin Cytoskeleton Signaling
ILK Signaling
Integrin Signaling
Cardiac Hypertrophy Signaling
RhoA Signaling
Leukocyte Extravasation Signaling
VEGF Signaling
CREB Signaling in Neurons
Signaling by Rho Family GTPases
Neuropathic Pain Signaling In Dorsal Horn Ne...
Gα12/13 Signaling
Paxillin Signaling
Regulation of Actin-based Motility by Rho
PAK Signaling
Tec Kinase Signaling
Corticotropin Releasing Hormone Signaling
Role of NFAT in Cardiac Hypertrophy
GNRH Signaling
© 2000-2016 QIAGEN. All rights reserved.
Canonical Pathway
Sorted by P-value:
-log(p-value)
0.00E00
13.9
Harderian
Lung
Thyroid
Liver
Calcium Signaling
Cellular Effects of Sildenafil (Viagra)
Actin Cytoskeleton Signaling
Protein Kinase A Signaling
Agranulocyte Adhesion and Diapedesis
Hepatic Fibrosis / Hepatic Stellate Cell Activat...
ILK Signaling
Epithelial Adherens Junction Signaling
GABA Receptor Signaling
Cell Cycle: G2/M DNA Damage Checkpoint R...
Serine Biosynthesis
Superpathway of Serine and Glycine Biosynth...
Histidine Degradation III
Xenobiotic Metabolism Signaling
Glutathione-mediated Detoxification
DNA damage-induced 14-3-3σ Signaling
LPS/IL-1 Mediated Inhibition of RXR Function
GADD45 Signaling
© 2000-2016 QIAGEN. All rights reserved.

In addition, transcriptomics data yielded benchmark dose (BMD; 95% lower confidence limits or BMDLs are shown) values that are in excellent agreement with the BMDLs derived from cancer endpoints. Testes data taken from (Recio et al., 2017).

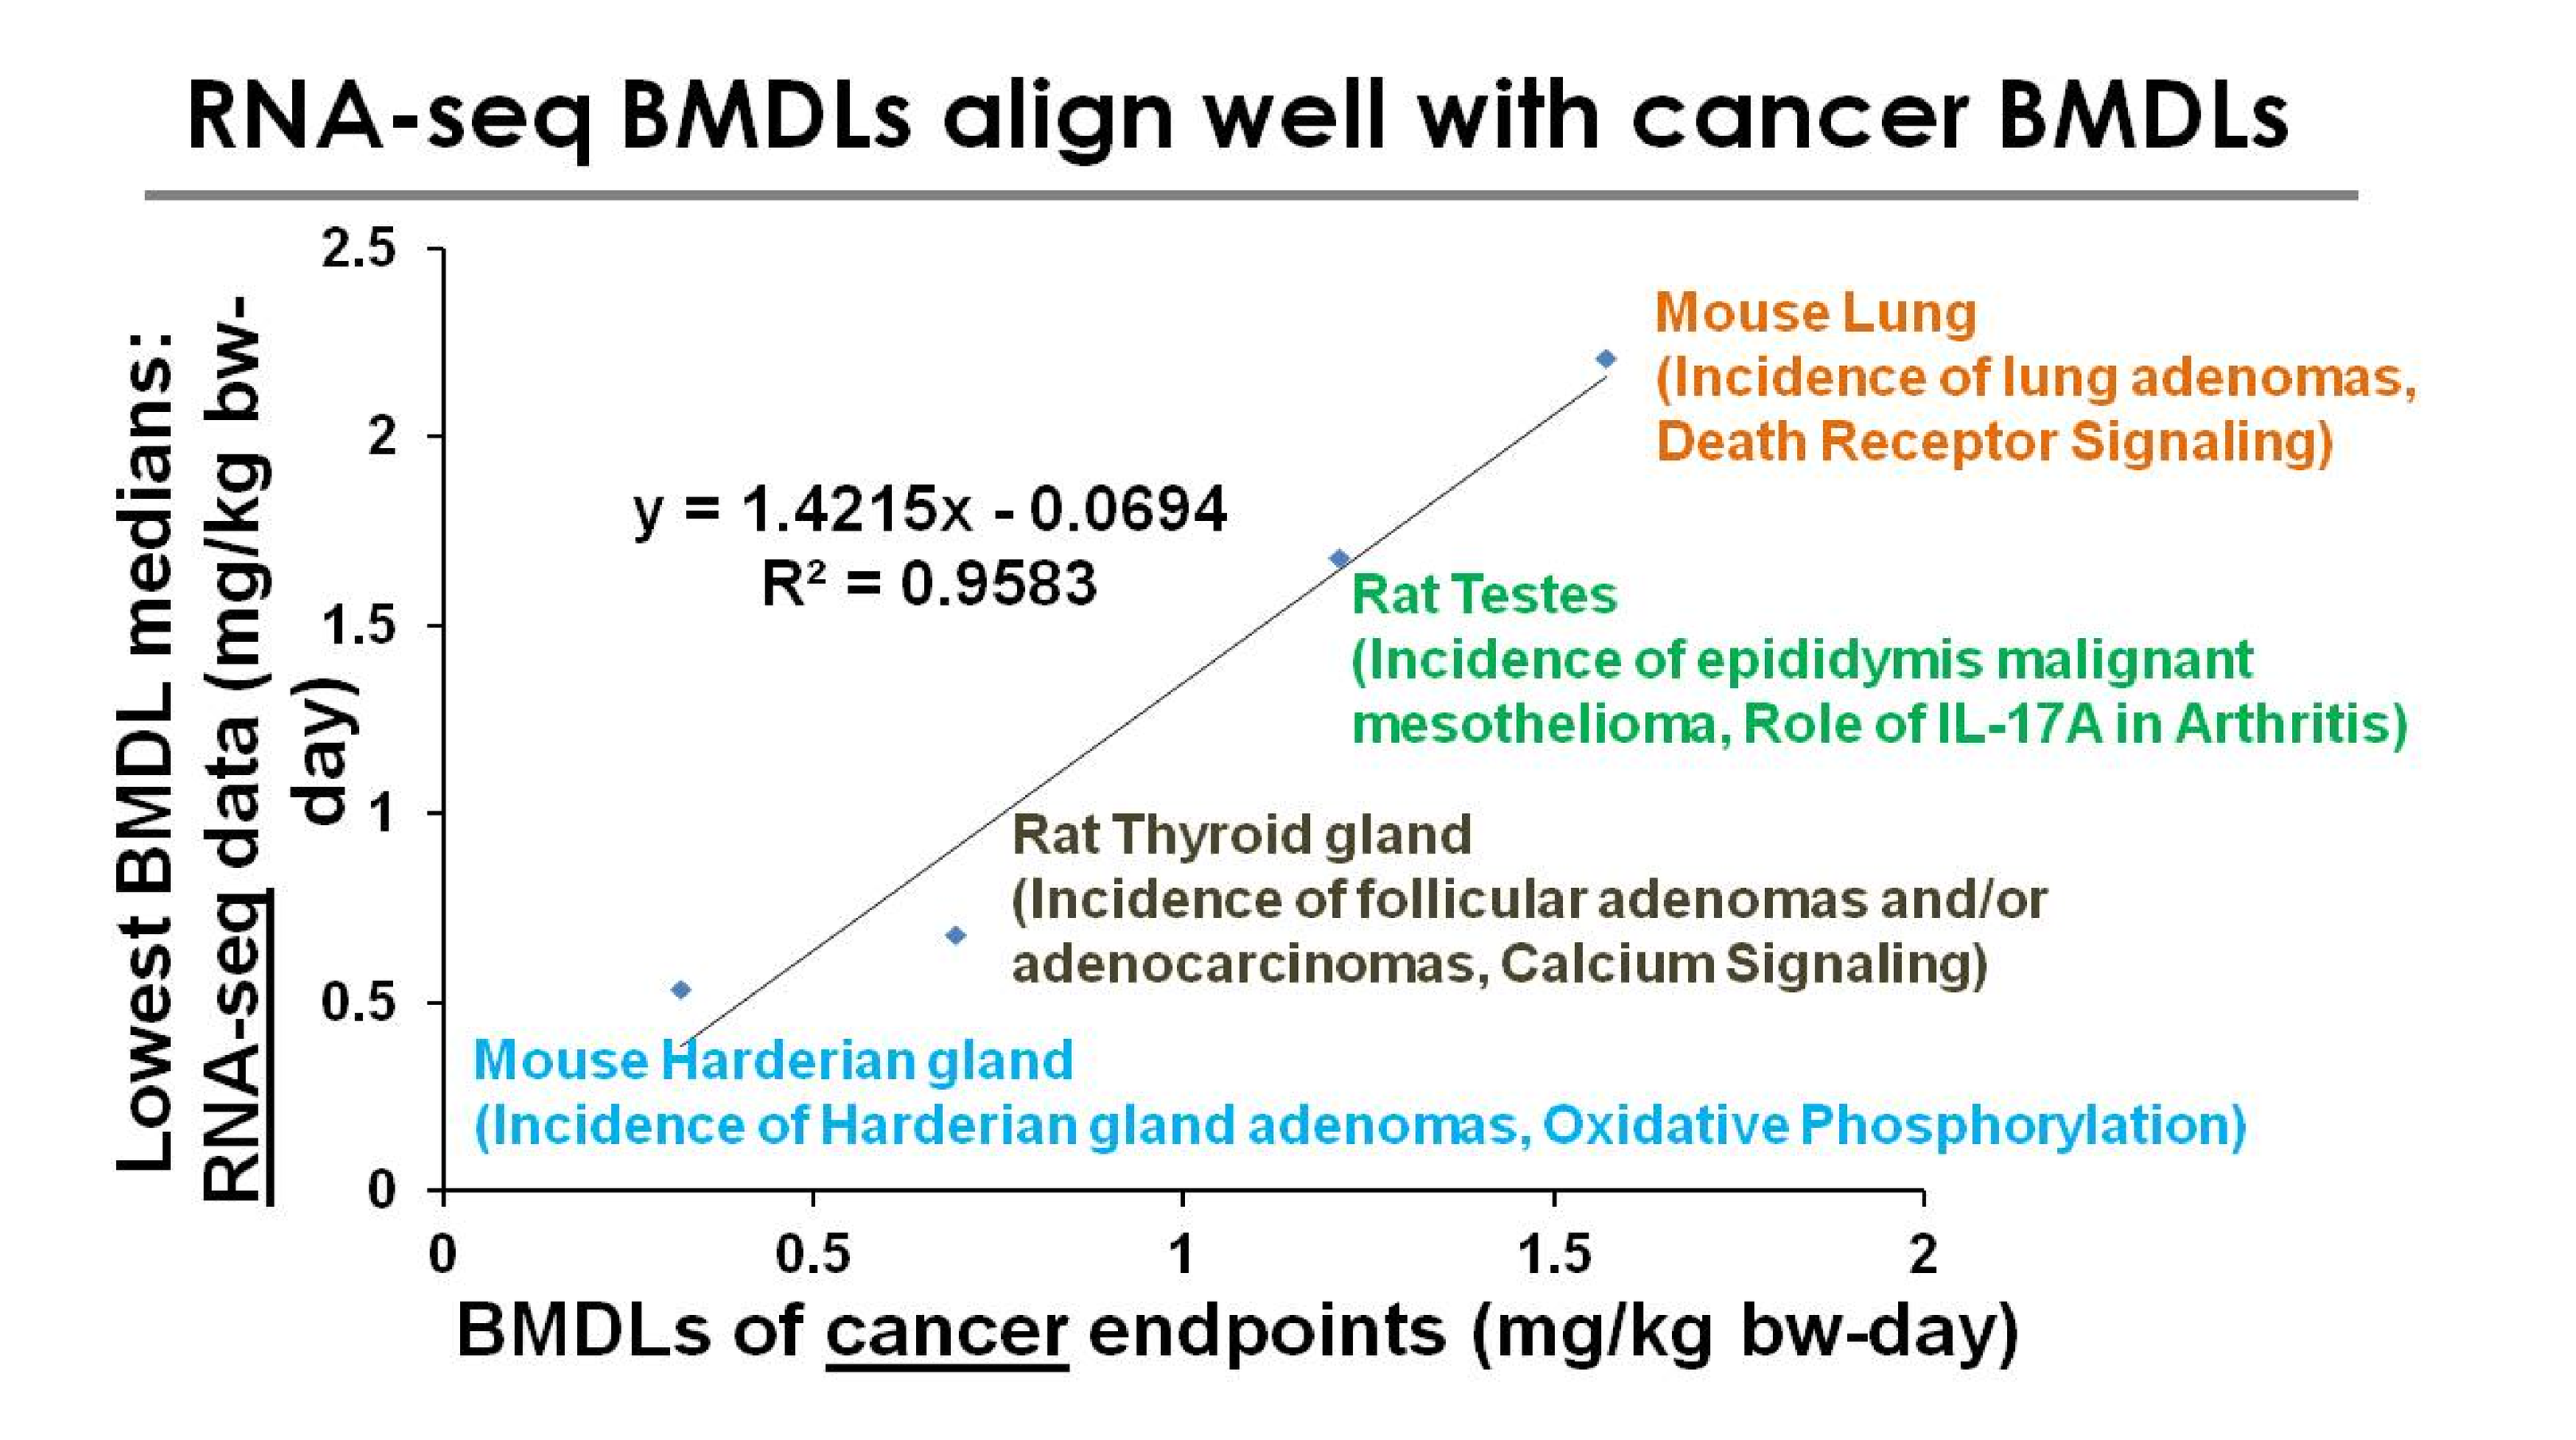
RNA-seq BMDLs align well with cancer BMDLs
Lowest BMDL medians: RNA-seq data (mg/kg bw-day)
BMDLs of cancer endpoints (mg/kg bw-day)
y = 1.4215x - 0.0694
R² = 0.9583
Mouse Lung (Incidence of lung adenomas, Death Receptor Signaling)
Rat Testes (Incidence of epididymis malignant mesothelioma, Role of IL-17A in Arthritis)
Rat Thyroid gland (Incidence of follicular adenomas and/or adenocarcinomas, Calcium Signaling)
Mouse Harderian gland (Incidence of Harderian gland adenomas, Oxidative Phosphorylation)
0
0.5
1
1.5
2
2.5

Discussion

● GENOTOXIC MOA:There was no effect of AA exposure on genes that are typically involved in DNA-damage response (i.e., such as those observed in animals exposed to genotoxic carcinogens that directly damage DNA, e.g., benzo[a]pyrene);

-this was consistent with a lack of induction of red blood cell micronuclei and equivocal results in the *Pig-a* mutation assay and **does not** support a genotoxic MOA;

●NOVEL, CA^{2+} SIGNALING-BASED MOA (CS MOA):

-Toxicogenomics provides **most support** for this MOA, including:

-reduced Ca^{2+} content in malignant tumors in post-mortem samples in humans (Beebe, 1904) and rodents (Clowes and Frisbie, 1905);

-effects of AA on genes associated with structural proteins, in accord with previous reports of AA binding to structural proteins (e.g. [Sickles *et al.*, 2007]);

-consistency with a similar MOA for AA-induced testicular toxicity (Recio et al., 2017);

In addition, RNA-seq data from the cancer target tissues of AA from mice and rats provide toxicogenomic BMDLs that are in excellent agreement with BMDLs from 2-year cancer bioassays.

Conclusions and Future Directions

This study provides an example of utility of toxicogenomics profiling to identify MOA and establish acceptable exposure levels.

This extensive study strongly suggests that analysis of calcium levels, associated signaling pathways and potential cytoskeletal effects, should be the subject of future studies on AA toxicity.

RNA-seq data from the cancer target tissues of AA from mice and rats provide toxicogenomic BMDLs that are in excellent agreement with BMDLs from 2-year cancer bioassays, once again supporting the utility of toxicogenomics in human health risk assessment.

Acknowledgements.

This study was supported by SNF SAS and Health Canada funds.

References

Recio, L. et al. (2017). Int. J. Toxicol., In press; Sickles, D. W., et al. (1996). J. Neurosci. Res. 46, 7-17; Beebe, P. (1904). Am. J. Physiol. 12, 167-172; Clowes, G. H. A. and Frisbie, W. S. (1905). Am. J. Physiol. 14, 173-192.